Case Report: Anaphylaxis from Western Harvester Ant

AUTHORS: M. Soteres, D. Soteres MD

ASTHMA & ALLERGY ASSOCIATES, PC AND RESEARCH CENTER

INTRODUCTION

Insect allergy is a global health issue that is often not identified and is a common subject of misunderstanding.[1] The Western Harvester Ant (WHA) (*Pogonomyrmex occidentalis*) is an endemic ant species to Colorado.[2] We are reporting a unique case of anaphylaxis from Western Harvester Ant (WHA). No other reports of WHA anaphylaxis have been identified.

CASE REPORT

A 13-year-old male with asthma and allergic rhinitis was conducting an insect identification project at school when stung by an ant. Using an insect identification application on his iPhone, he identified the ant as a WHA. The reaction started as large local erythema and his mother took him to the emergency room.
At the ER, symptoms progressed to throat tightening, dizziness, and wheezing. He was treated with antihistamines, epinephrine, and albuterol, with good response. He had a virtual consult at Anschutz five months after the event and labs were ordered.

- Tryptase = 2.8 µg/L (Normal: ≤ 10.9 µg/L)
- Total IgE = 1368 kuA/L (Normal: 0.0 - 87.0 kuA/L)
- Fire Ant IgE = 7.2 kU/L (Normal: <0.70 kU/L)

He came to our clinic with his mother for further evaluation and treatment options.

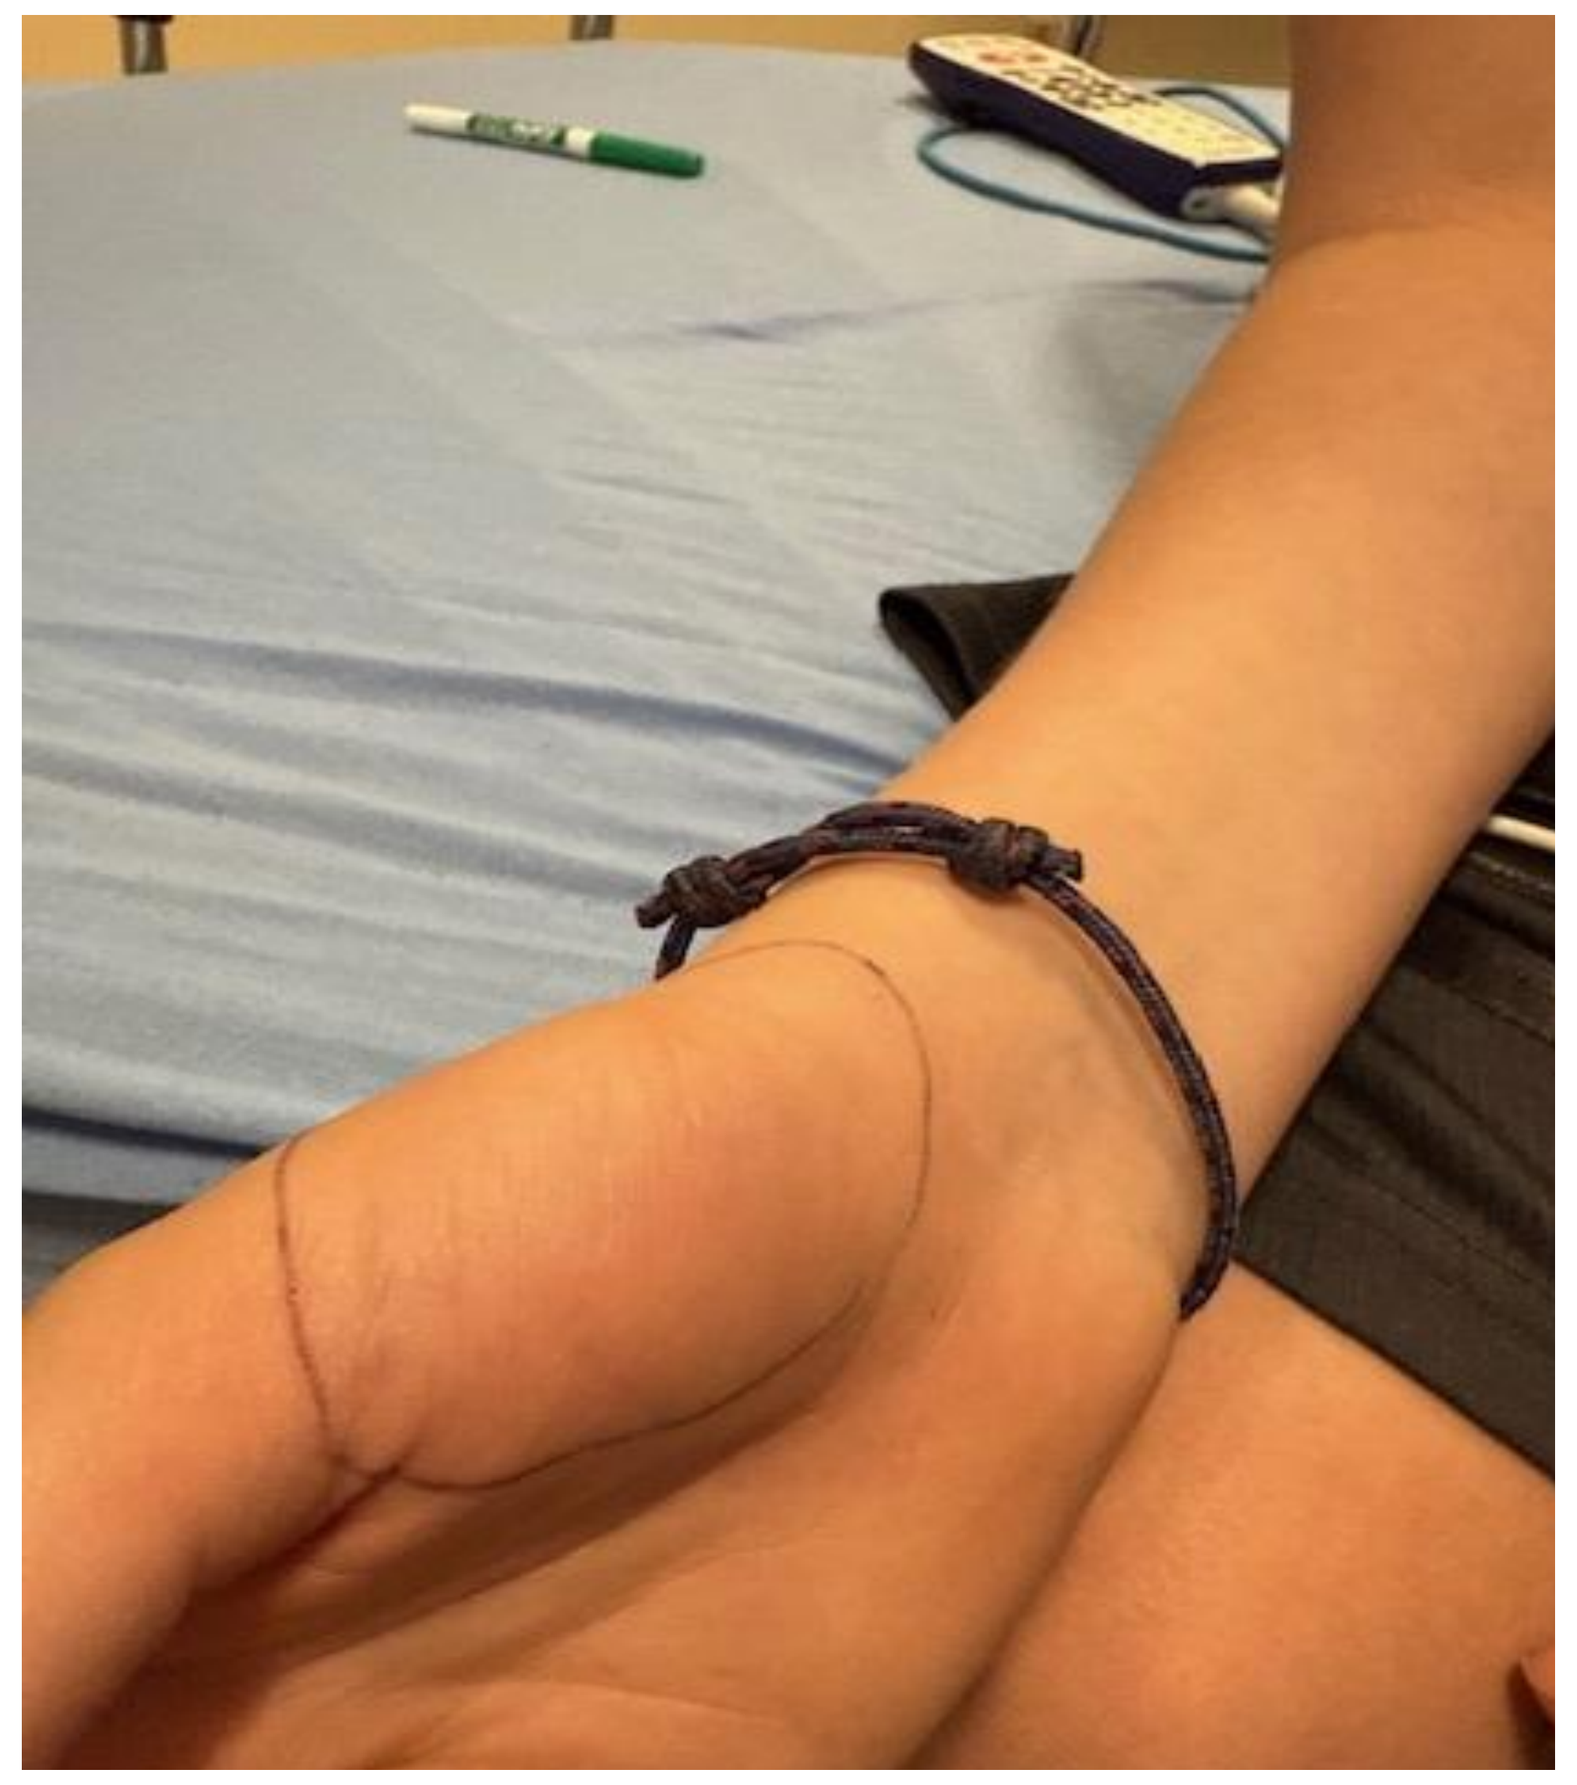

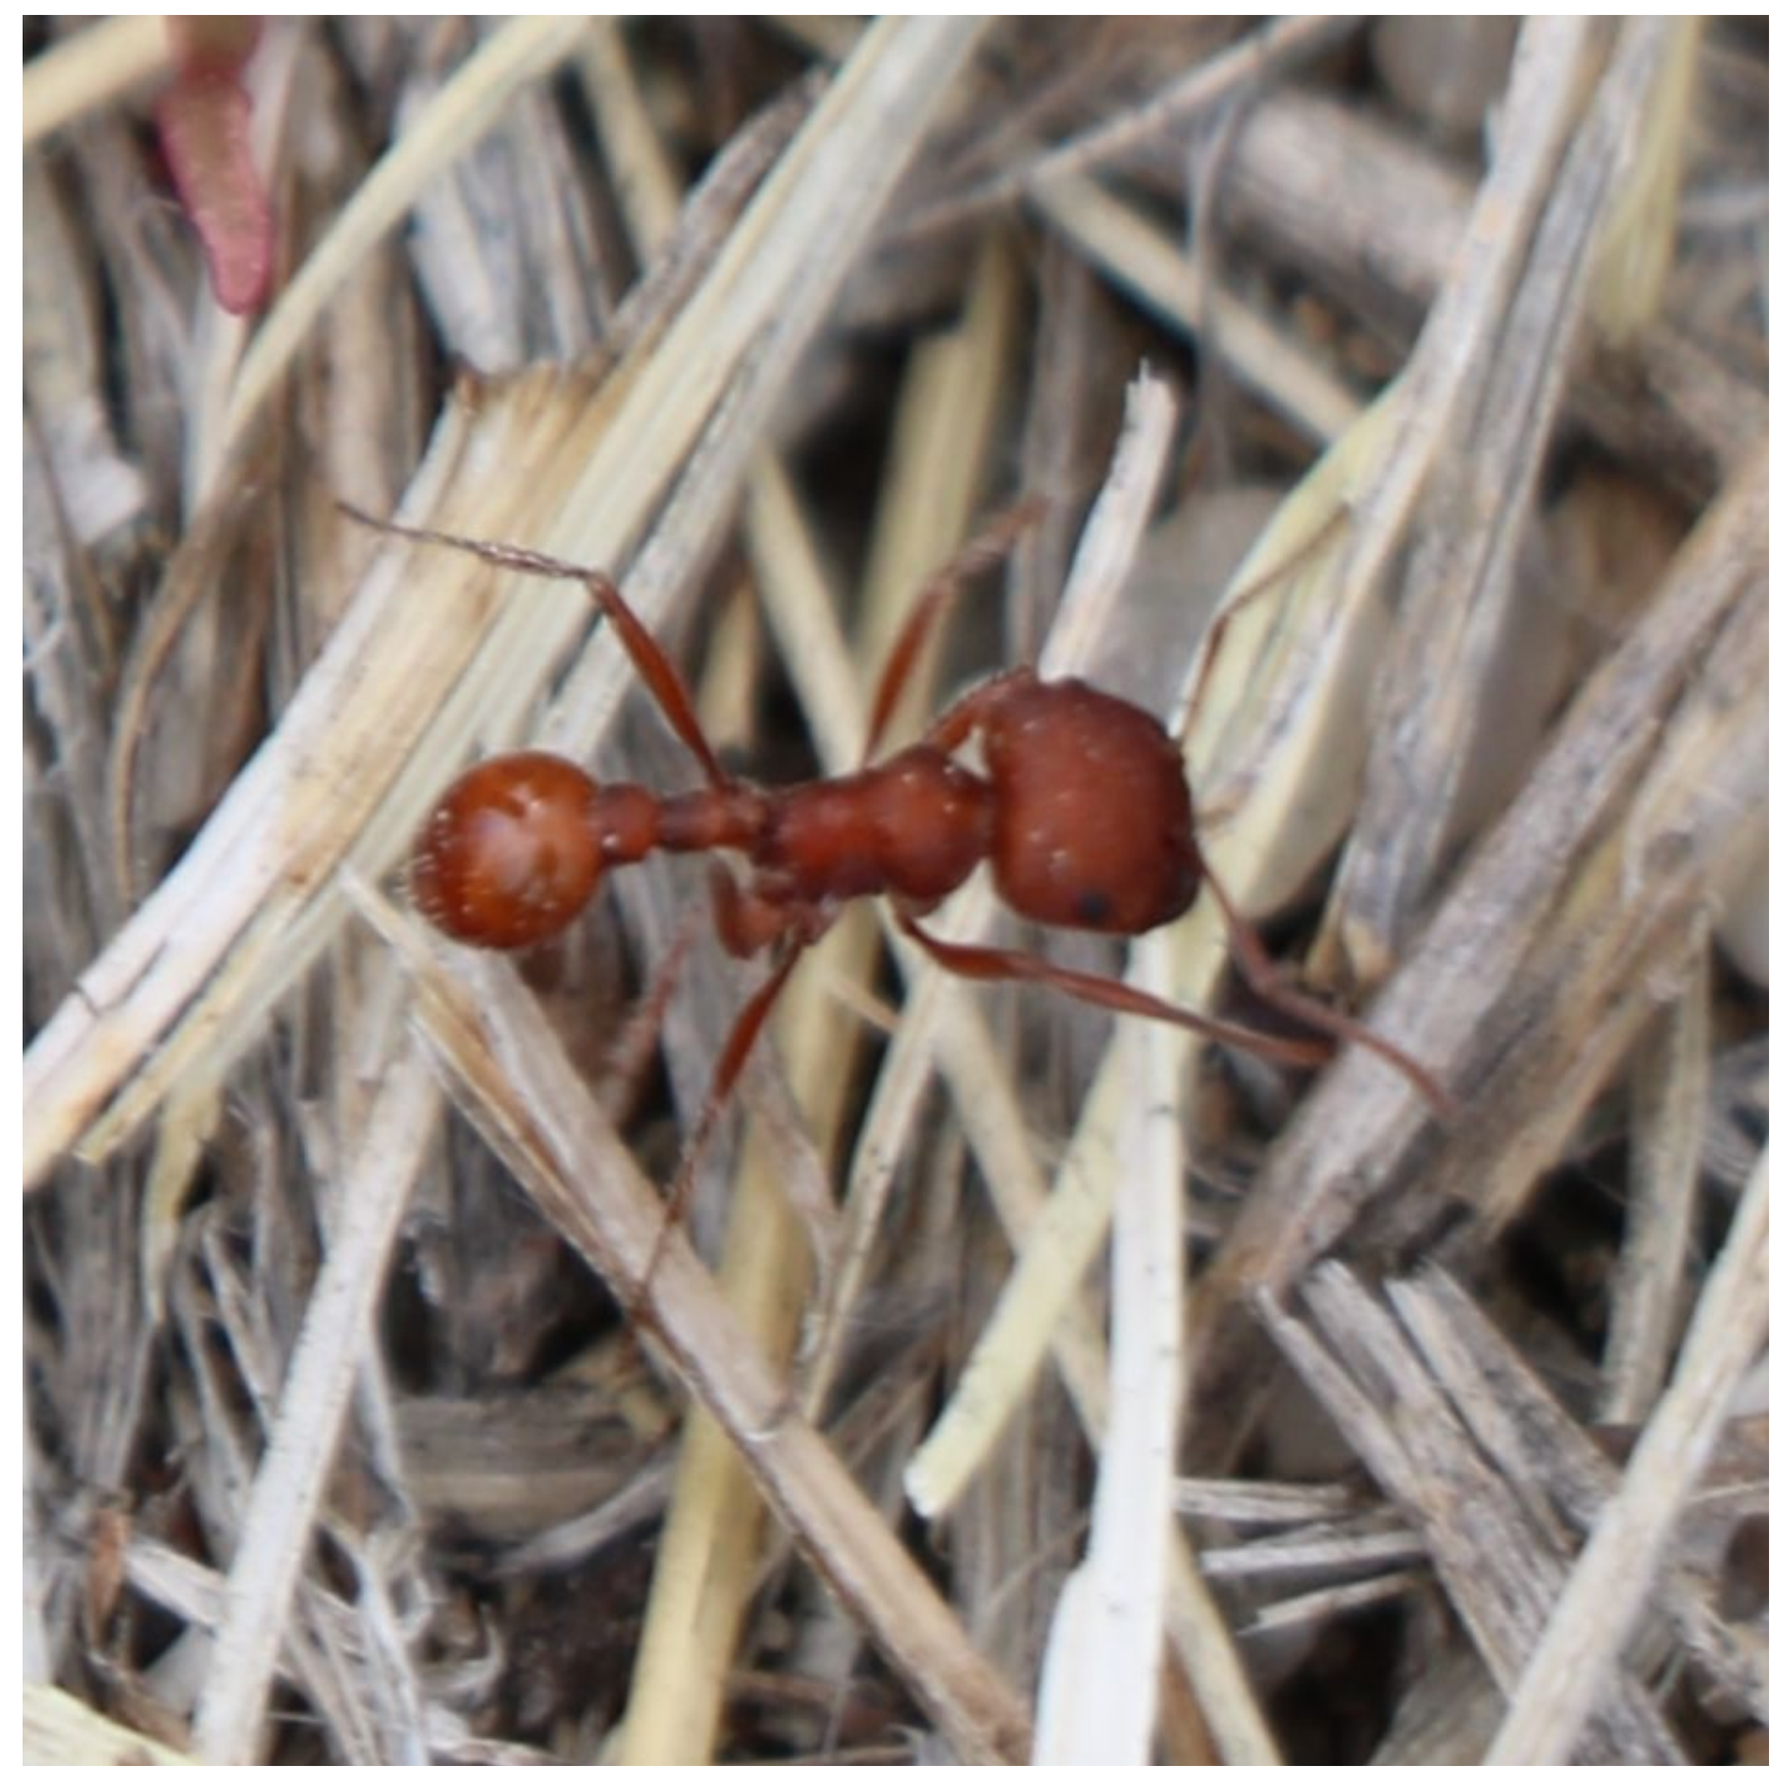

REFERENCES

1. Golden, David B. K. "15 - Insect Allergy." In *Allergy Essentials (Second Edition)*. Elsevier, 2022. https://doi.org/10.1016/B978-0-323-80912-2.00015-9.
2. iNaturalist. "Western Harvester Ant (Pogonomyrmex Occidentalis)." Accessed June 29, 2026. https://www.inaturalist.org/taxa/69514-Pogonomyrmex-occidentalis.
3. Pinnas, J. L., R. C. Strunk, T. M. Wang, and H. C. Thompson. "Harvester Ant Sensitivity: In Vitro and in Vivo Studies Using Whole Body Extracts and Venom." *The Journal of Allergy and Clinical Immunology* 59, no. 1 (1977): 10–16. https://doi.org/10.1016/0091-6749(77)90170-1.
4. Weber, Richard W. "Allergen of the Month—Harvester Ant." *Annals of Allergy, Asthma & Immunology* 111, no. 3 (2013): A19. https://doi.org/10.1016/j.anai.2013.07.021.
5. Keeler, Kathleen H. "LONGITUDINAL STUDY OF POGONOMYRMEX OCCIDENTALIS (HYMENOPTERA: FORMICIDAE), WESTERN HARVESTER ANT, COLONY SURVIVAL IN WESTERN NEBRASKA." *The Southwestern Naturalist* 66, no. 2 (2022). https://doi.org/10.1894/0038-4909-66.2.114.
6. Academy of Natural Sciences of Philadelphia., Academy of Natural Sciences of Philadelphia, and American Entomological Society. *Entomological News*. V.70 (1959) [Lacks:pg.114-115]. American Entomological Society, 1959. https://www.biodiversitylibrary.org/item/20697.
7. Schmidt, J. O., and M. S. Blum. "A Harvester Ant Venom: Chemistry and Pharmacology." *Science* (New York, N.Y.) 200, no. 4345 (1978): 1064–66. https://doi.org/10.1126/science.653354.
8. McMurray, Jeremy C., Karla E. Adams, Troy Wanandy, Adriana Le, and Robert J. Heddle. "Stinging Ant Anaphylaxis: Advances in Diagnosis and Treatment." *The Journal of Allergy and Clinical Immunology. In Practice* 13, no. 1 (2025): 25–37. https://doi.org/10.1016/j.jaip.2024.07.014.

HISTORY

- *P. occidentalis* is widespread throughout the Western United States.[3]
- Nests are fully exposed to the sun[3] and the surrounding area (referred to as the disc) is cleared of both dead and living vegetation.[5]
- Construction of nests takes places in sand or soil,[3] with a distinct single opening.[4]
- In Colorado, the discs of mature colonies average 1.4 m^2.[5]
- Colonies are founded by a single, multiple-mating queen who lives an average of 16-19 years.[5]
- The average survival length for a colony is 16-19 years.[5]
- When observed, *Pogonomyrmex* ants bite with their mandibles, then pivot and sting multiple times in a circular motion, similar to the better-known *Solenopsis* fire ant sting.[4]
- The last publication on *P. occidentalis* stings was Neal Weber's 1959 report in *Entomological News* on six cases of painful, local reactions that occurred in 1942.[6]

VENOM

Pogonomyrmex venom is not cross-reactive with other insect venoms, including that of *Solenopsis*.[3] Currently, there is no research on the components of WHA venom; however, a study conducted by Jacob Pinnas suggests that *Pogonomyrmex* harvester ant sensitivity is genus-specific.[3] This was demonstrated through a comparison of *P. rugosus* and *P. mariposa*. Pinnas found that rabbits immunized with whole-body extracts made from either species developed precipitating antibodies against the immunization extract. These antibodies cross-reacted in a line of identity against another *Pogonomyrmex* species. It did not react to Formica ant, fire ant, carpenter ant, or a mixture derived from bee, wasp, yellow jacket, and hornet.[3]

Additionally, current research suggests that Harvester Ant venom may pose a greater risk for allergic reactions than other insect venom. *P. badius* particularly has a higher potency algogenic venom. The direct hemolytic activity is not reduced by incubation of the venom with a 100-fold excess of heparin.[7] This raises the prospect that heparin lacks the ability to form complexes with harvester ant venom or to reduce its activity. Their venom also contains elevated activity of phospholipase A and phospholipase B, along with a high concentration of lipase (which has not previously been reported in insect or animal venom).[7] Acid phosphatase and esterase enzymes are also present in harvester ant venom, although they have not been observed previously in ant venom.[7] It is important to consider that these enzymes, which are elevated in harvester ant venom, are major allergens, meaning that harvester ant venom may pose a greater risk for allergic reactions and anaphylaxis than previously thought.[7]

SPECIES RANGE MAP: P. OCCIDENTALIS

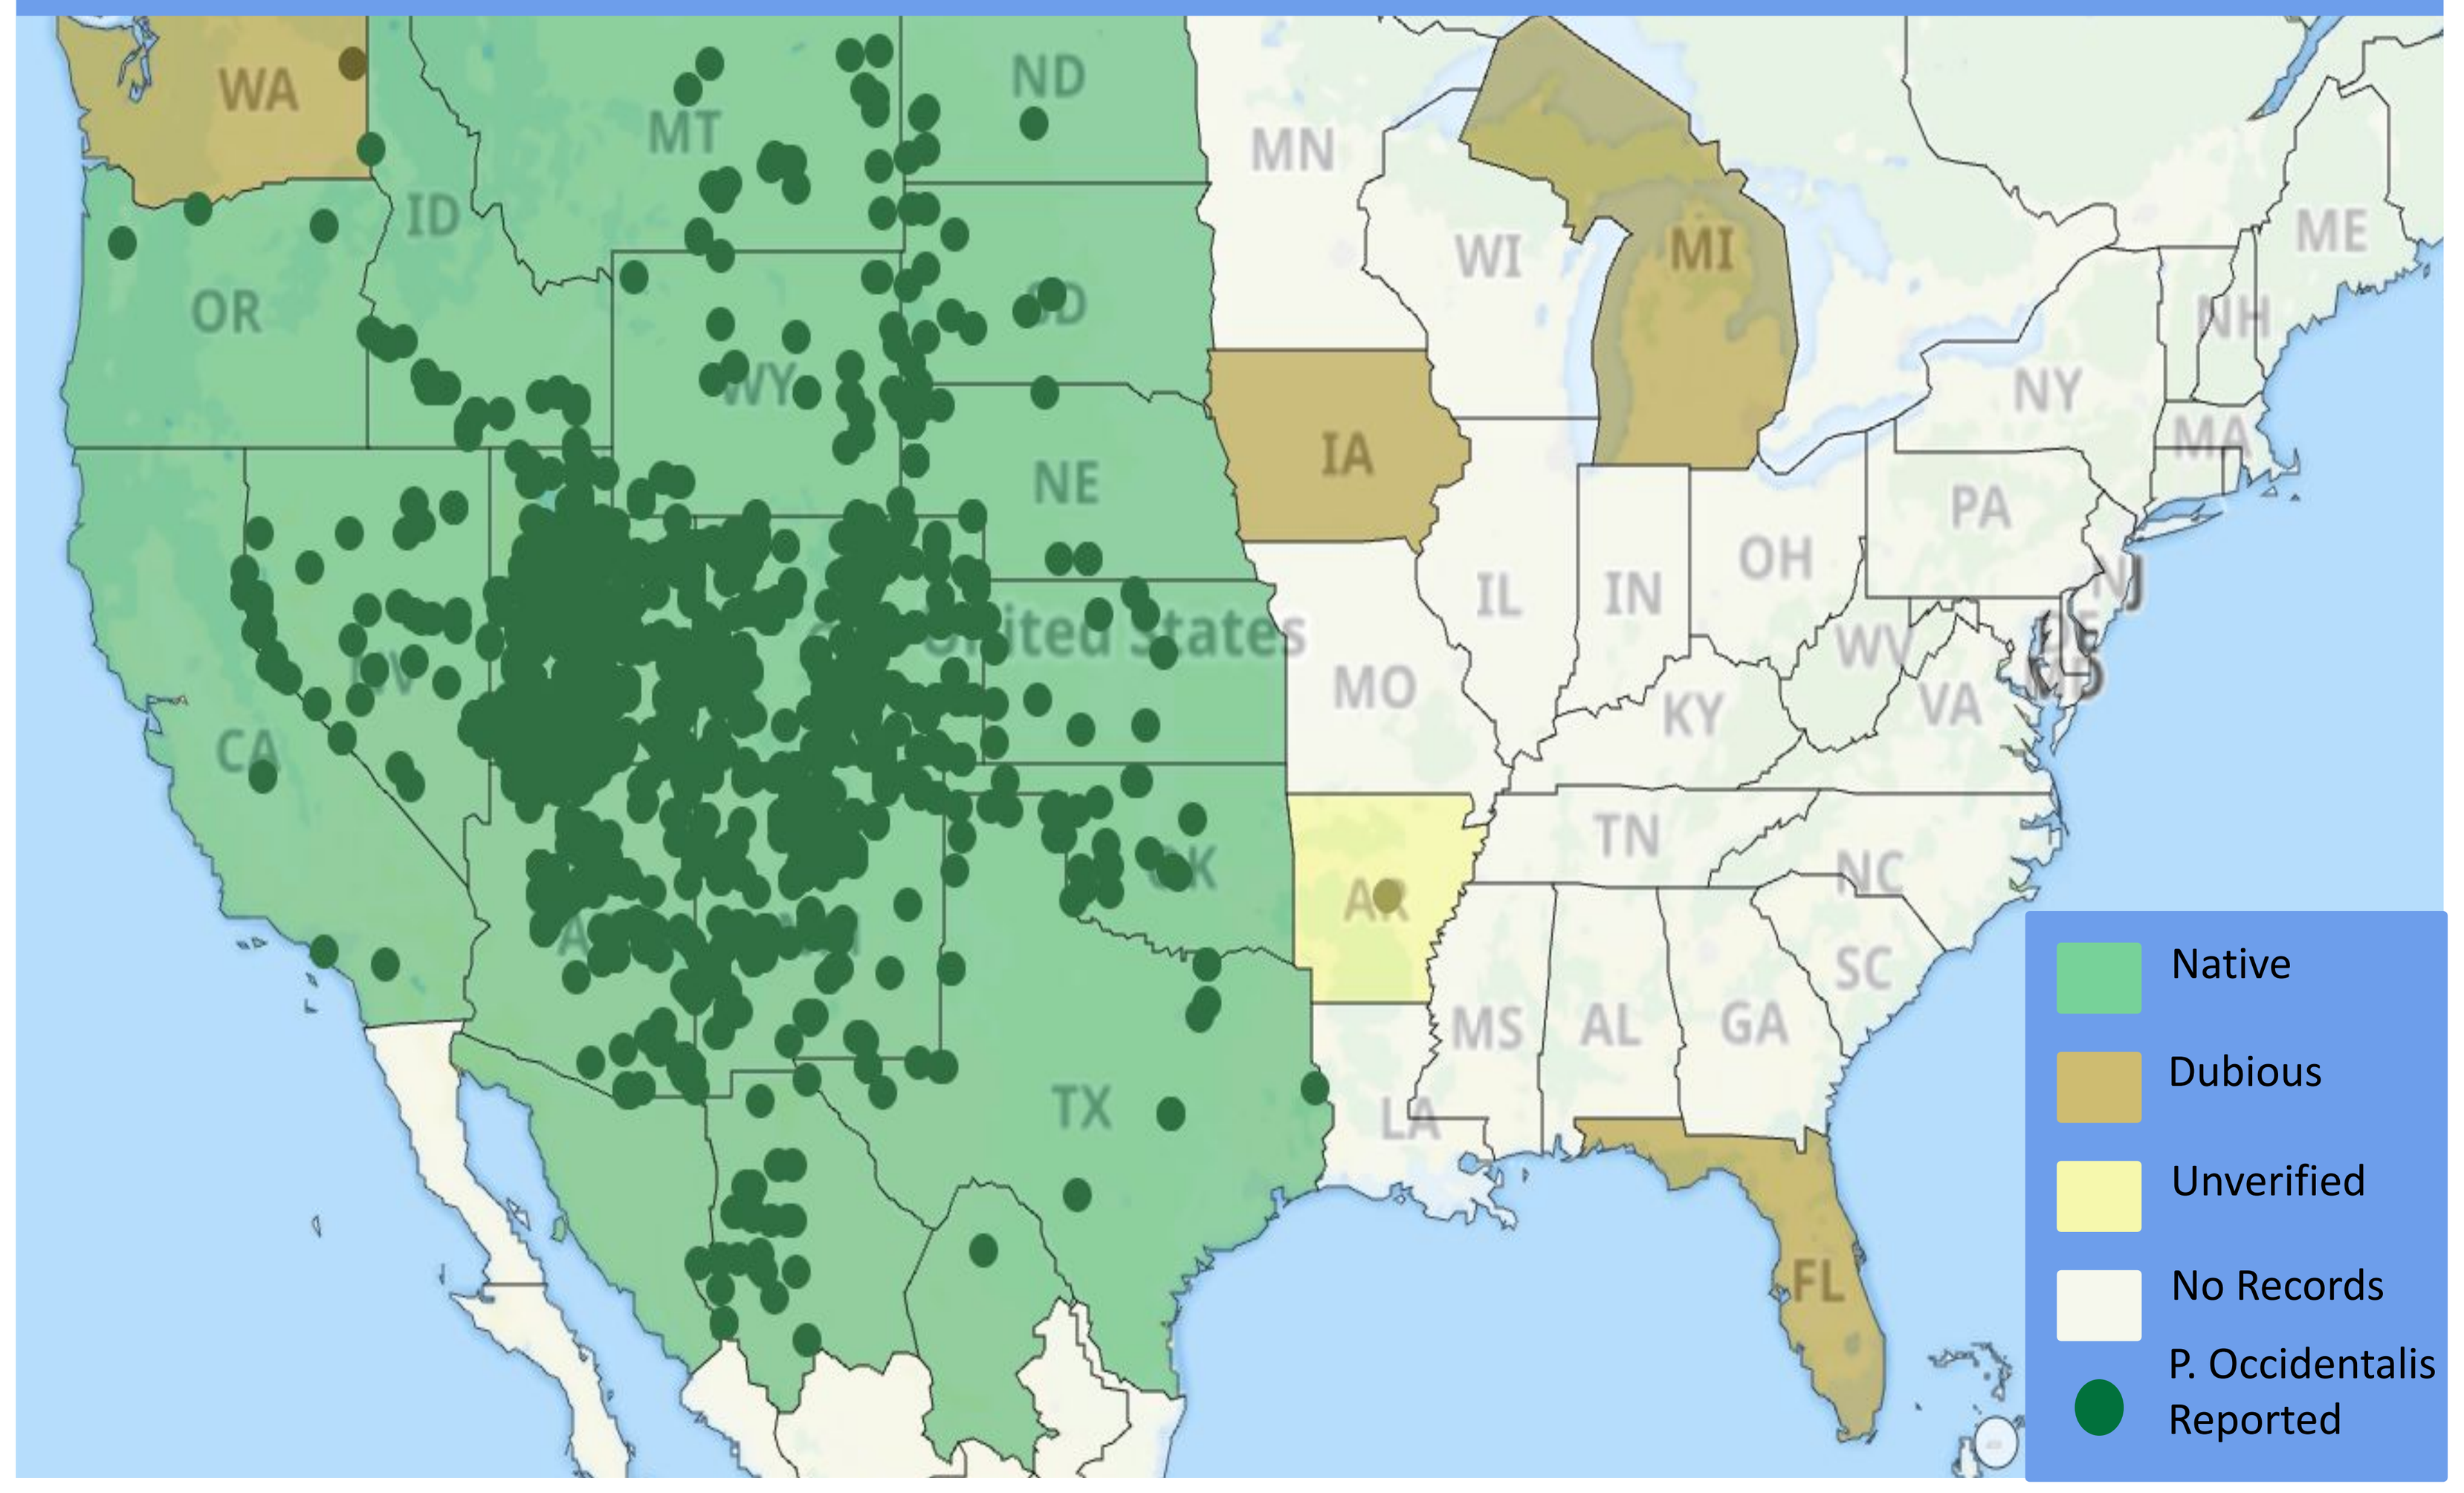

WA
OR
CA
ID
MT
WY
ND
NE
TX
MN
WI
MI
IA
IL
IN
OH
MO
KY
TN
NC
SC
MS
AL
GA
FL
PA
NY
ME
Native
Dubious
Unverified
No Records
P. Occidentalis Reported

Guénard, B., Weiser, M., Gomez, K., Narula, N., Economo, E.P. (2017) The Global Ant Biodiversity Informatics (GABI) database: a synthesis of ant species geographic distributions. Myrmecological News 24: 83-89. Janicki, J., Narula, N., Ziegler, M., Guénard, B. Economo, E.P. (2016) Visualizing and interacting with large-volume biodiversity data using client-server web-mapping applications: The design and implementation of antmaps.org. Ecological Informatics 32: 185-193

CASE MANAGEMENT

- Patient and family educated about risk of worsening reaction with repeat bite.
- Discussed importance of recognizing risk and measures to reduce exposure.
- Educated on signs and symptoms of anaphylaxis and treatment algorithms using injectable or other epinephrine and oral benadryl.
- Reviewed option of Fire Ant Immunotherapy due to abnormal labs; however, he had never been exposed to fire ant and does not live in endemic region. Additionally, current scientific knowledge suggests no cross reactivity between WHA and fire ant.[3]

DISCUSSION

- Our literature search for cases of WHA systemic allergic reactions or anaphylaxis yielded no other reports.
- Currently, In vitro testing is only commercially available for *Solenopsis invicta*, known as the red imported fire ant or IFA, and *Myrmecia pilosula*, known as the jack jumper ant or JJA.[8] We are unable to perform specific testing for WHA sensitivity.
- Our goal is to raise awareness of the possibility of WHA anaphylaxis for endemic regions.
- Future endeavors should be considered to develop and standardize testing for Western Harvester Ant Anaphylaxis.